TG02, an oral CDK inhibitor, induces caspase-independent, non-autophagic cell death in human glioma cell lines and glioma-initiating cells

Emilie Le Rhun[1,2], Caroline Von Achenbach[1], Emese Szabo[1] and Michael Weller[3]
(1) Department of Neurology, University Hospital and University of Zurich, Zurich, Switzerland, (2) University Hospital Lille, Lille, France

Background

TG02 is an orally bioavailable, multikinase inhibitor which potently inhibits cyclin-dependent kinases (CDK) 1, 2, 5, 7 and 9. CDK9-dependent depletion of short-lived oncoproteins such as MCL-1 and MYC has been proposed as its primary mechanism of cytotoxicity.

TG02 target gene expression is not related to outcome in glioblastoma: an analysis of the TCGA database

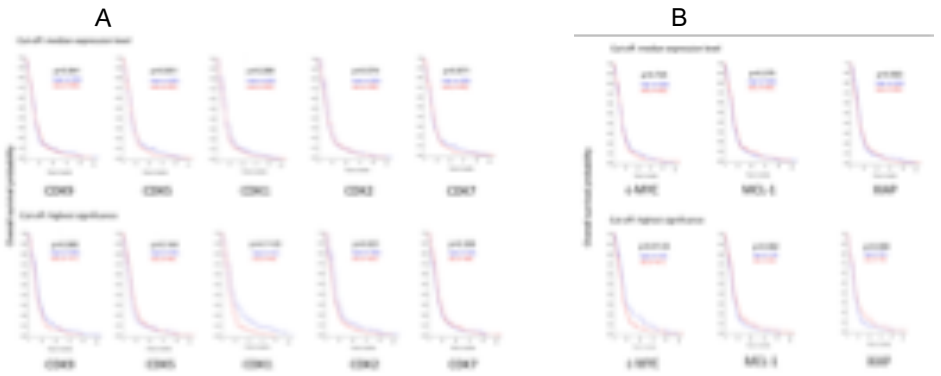
A
B

Kaplan-Meier survival curves of overall survival are shown for newly diagnosed glioblastoma patients. Patients were divided into two groups with high (blue) or low (red) expression of the direct kinase targets, CDK 1, 2, 5, 7 or 9 (A), or the indirect targets MYC, MCL-1 and XIAP (B), based either on median mRNA expression level (upper row) or expression level that results in the highest association with survival (lower row). Statistical significances (p) were determined using the log-rank test (p<0.05 was considered significant).

Single TG02 target gene expression does not predict TG02 sensitivity

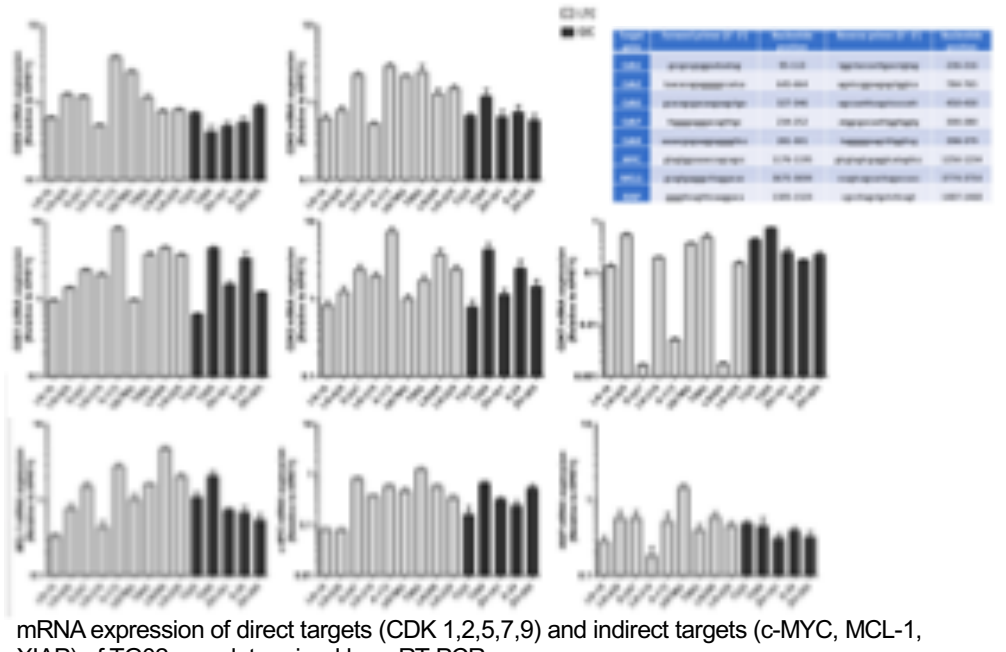

mRNA expression of direct targets (CDK 1,2,5,7,9) and indirect targets (c-MYC, MCL-1, XIAP) of TG02 was determined by q-RT-PCR.

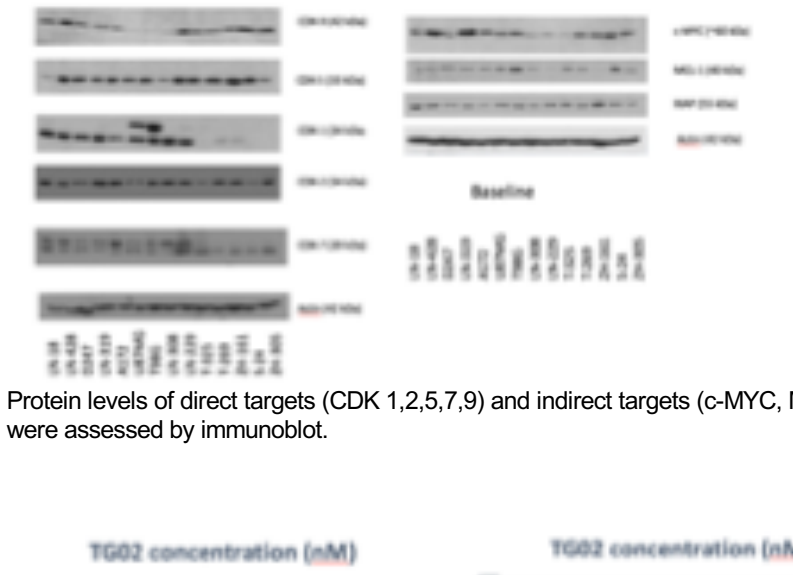
Baseline

Protein levels of direct targets (CDK 1,2,5,7,9) and indirect targets (c-MYC, MCL-1, XIAP) were assessed by immunoblot.

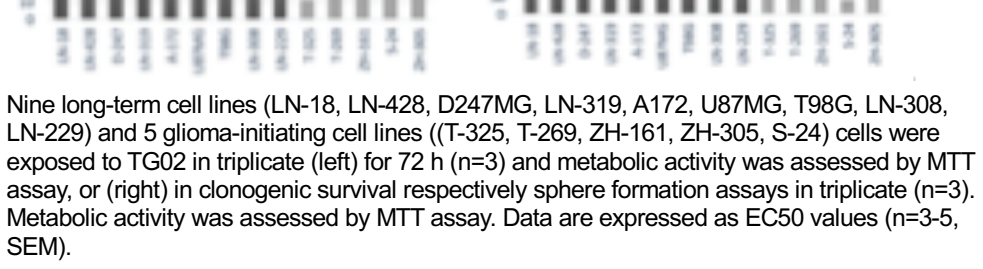
TG02 concentration (nM)
TG02 concentration (nM)

Nine long-term cell lines (LN-18, LN-428, D247MG, LN-319, A172, U87MG, T98G, LN-308, LN-229) and 5 glioma-initiating cell lines ((T-325, T-269, ZH-161, ZH-305, S-24) cells were exposed to TG02 in triplicate (left) for 72 h (n=3) and metabolic activity was assessed by MTT assay, or (right) in clonogenic survival respectively sphere formation assays in triplicate (n=3). Metabolic activity was assessed by MTT assay. Data are expressed as EC50 values (n=3-5, SEM).

Confirmation of target gene suppression by TG02

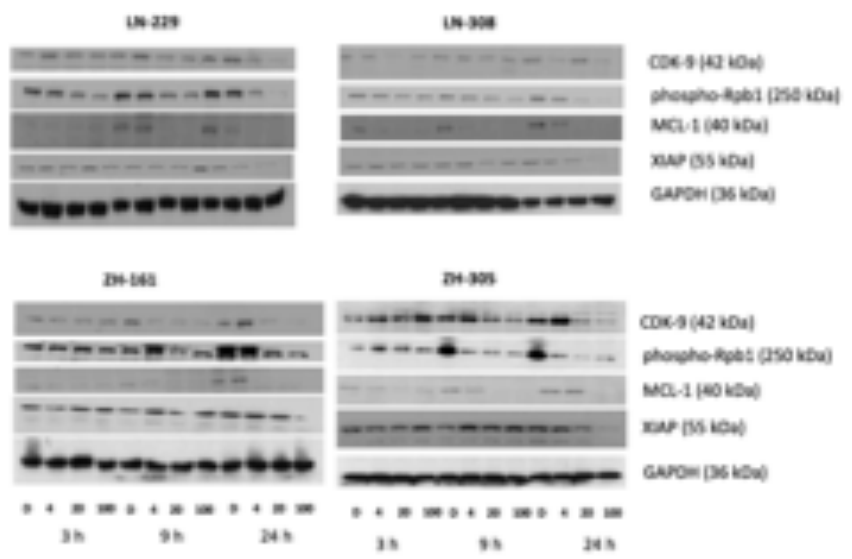
LN-229
LN-308
CDK-9 (42 kDa)
phospho-Rpb1 (250 kDa)
MCL-1 (40 kDa)
XIAP (55 kDa)
GAPDH (36 kDa)
ZH-161
ZH-305
3 h
9 h
24 h

LN-229, LN-308, ZH-161 or ZH-305 cells were exposed to TG02 at 4, 20 or 100 nM for 3, 9 or 24 h. Protein levels of CDK-9, phospho-Rpb1, MCL-1 and XIAP were determined by immunoblot.

No specific cell cycle alteration, but increased Anx labeling suggestive of apoptosis in response to TG02

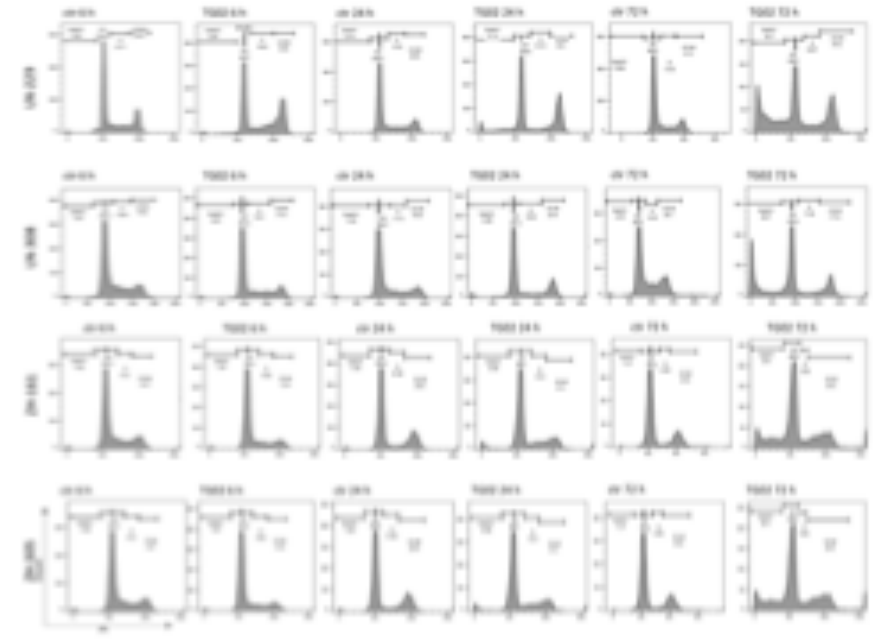

LN-229, LN-308, ZH-161 or ZH-305 cells were exposed to 100 nM TG02 or solvent control for 6, 24 or 72 h, permeabilized, stained with propidium iodide (PI), and then analysed for cell cycle distribution by flow cytometry.

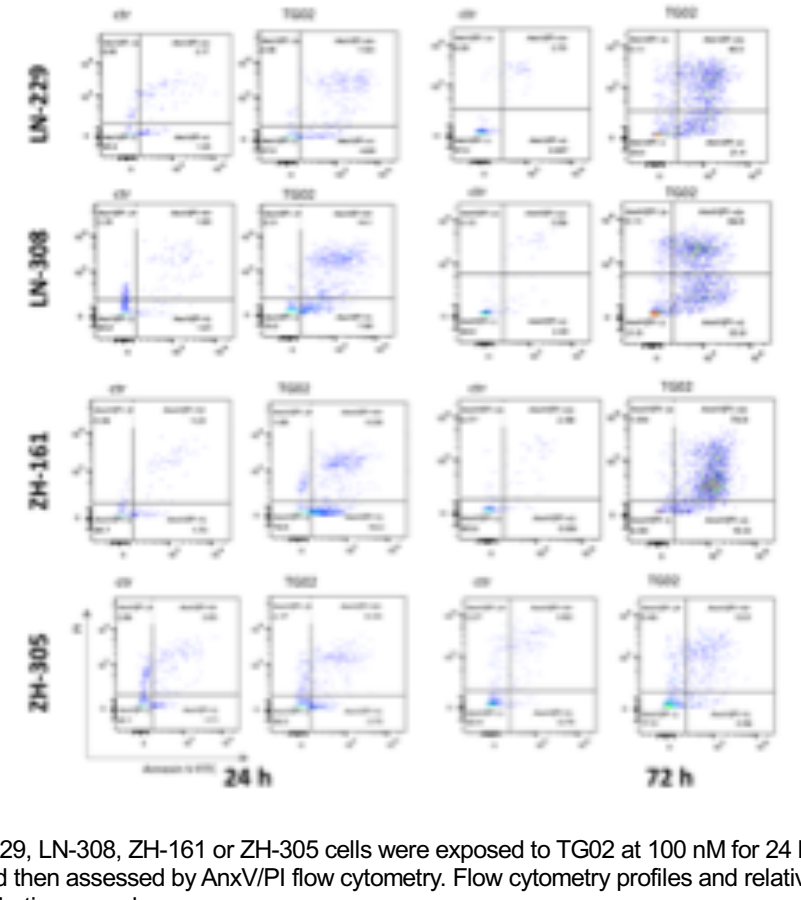
LN-229
LN-308
ZH-161
ZH-305
24 h
72 h

LN-229, LN-308, ZH-161 or ZH-305 cells were exposed to TG02 at 100 nM for 24 h or 72 h and then assessed by AnxV/PI flow cytometry. Flow cytometry profiles and relative distributions are shown.

Minor caspase 3 processing and a protective role of autophagy?

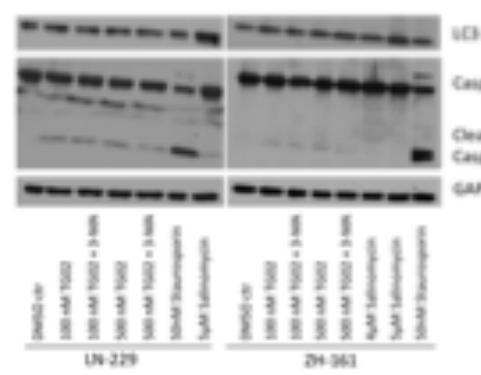
LC3 A/B I
Caspase 3
Cleaved Caspase 3
GAPDH
LN-229
ZH-161

Cells treated accordingly were assessed for caspase 3 or LC3 protein patterns by immunoblot. Parallel cultures were pretreated (1 h) and cotreated with 3-MA (1 mM). High concentrations of TG02 induced minor caspase 3 cleavage that was attenuated by 3-MA.

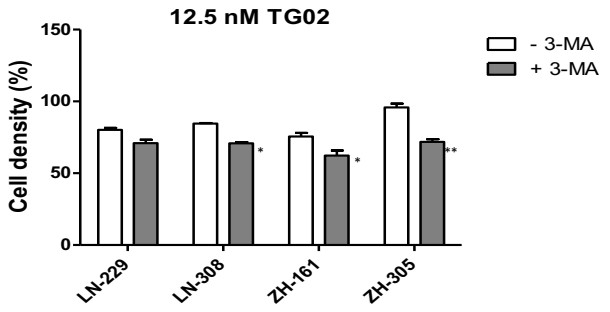
12.5 nM TG02
- 3-MA
+ 3-MA
Cell density (%)
150
100
50
0
LN-229
LN-308
ZH-161
ZH-305

The cells were pretreated or not for 1 h with 3-methyladenine (1 mM) and cotreated with TG02 for 72 h. 3-MA treatment enhanced growth inhibition mediated by TG02, suggesting that autophagy is protective.

Hypoxia confers resistance to TG02 to GIC cultures, but not to LTC cultures

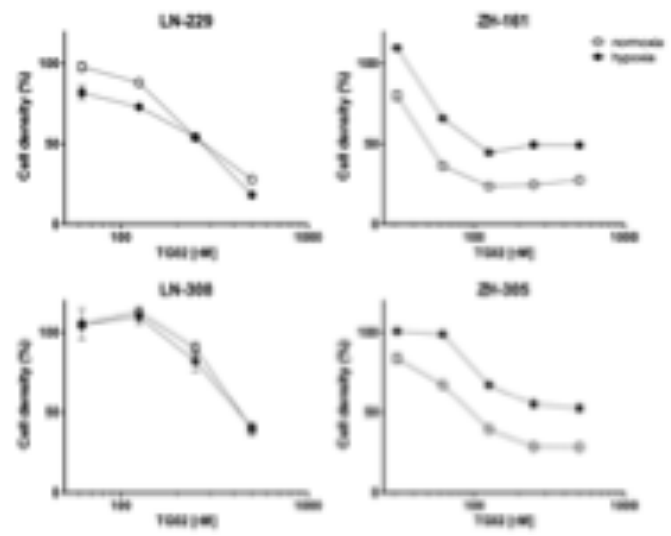
LN-229
ZH-161
LN-308
ZH-305

Under hypoxic cell culture conditions, TG02 was less active against the glioma-initiating cell lines, ZH-161 and ZH-305, but not against the long-term cell lines, LN-229 and LN-308.

Conclusions

Baseline expression of direct TG02 targets (CDK-9, CDK-5) and indirect TG02 targets (MCL-1, MYC) is detected in all cell lines by immunoblot, but does not correlate with sensitivity to TG02.

TG02 exhibits strong anti-tumor activity in both human long-term cell lines (LN-18, LN-428, D247MG, LN-319, A172, U87MG, T98G, LN-308, LN-229) and in glioma-initiating cell lines (T-325, T-269, ZH-161, ZH-305, S-24) regardless of MGMT promoter methylation status (not shown).

TG02 is a highly potent anti-glioma agent *in vitro* with a novel mode of action that induces cell death in a largely caspase-independent, non-autophagic manner. Further cell death studies are ongoing.

Early clinical trials of TG02 in glioblastoma are ongoing.